Molecular Characterization of VEGF Secretion by MSC-NTF Cells (NurOwn®): Therapeutic Implications in ALS

Haggai Kaspi, Revital Aricha, Natalie Abramov, Yael Gothelf, Chaim Lebovits, Joseph Petroziello, Yossef S. Levi, Ralph Kern

BrainStorm Cell Therapeutics, Petach Tikvah, Israel and New York City, NY, USA

Background

MSC-NTF cells (NurOwn®) are autologous bone marrow derived mesenchymal stem cells (MSC) induced in culture to secrete high levels of neurotrophic factors (NTFs) that support neuronal growth and survival. Thus, MSC-NTF cells combine MSC's immunomodulatory therapeutic benefits with enhanced neurotrophic factor secretion. A phase 3 randomized clinical trial is in progress at 6 sites in the US to evaluate the efficacy and safety of repeated intrathecal administration of MSC-NTF cells in ALS patients. As previously reported, VEGF has an important immunomodulatory and neuroprotective role in ALS through biological effects on microglia and glutamatergic disease mechanisms.

Methods

To explore the molecular differences between naïve MSC and MSC-NTF cells, we performed mass-spectrometry proteomics, Affymetrix gene array and microRNA array. To map changes in protein secretion, ELISA assays were employed. Subsequently, several bioinformatic tools were applied to investigate the interactions between protein secretion, protein/gene expression and microRNAs.

Results

Increased secretion of multiple NTFs by MSC-NTF cells *in-vitro*

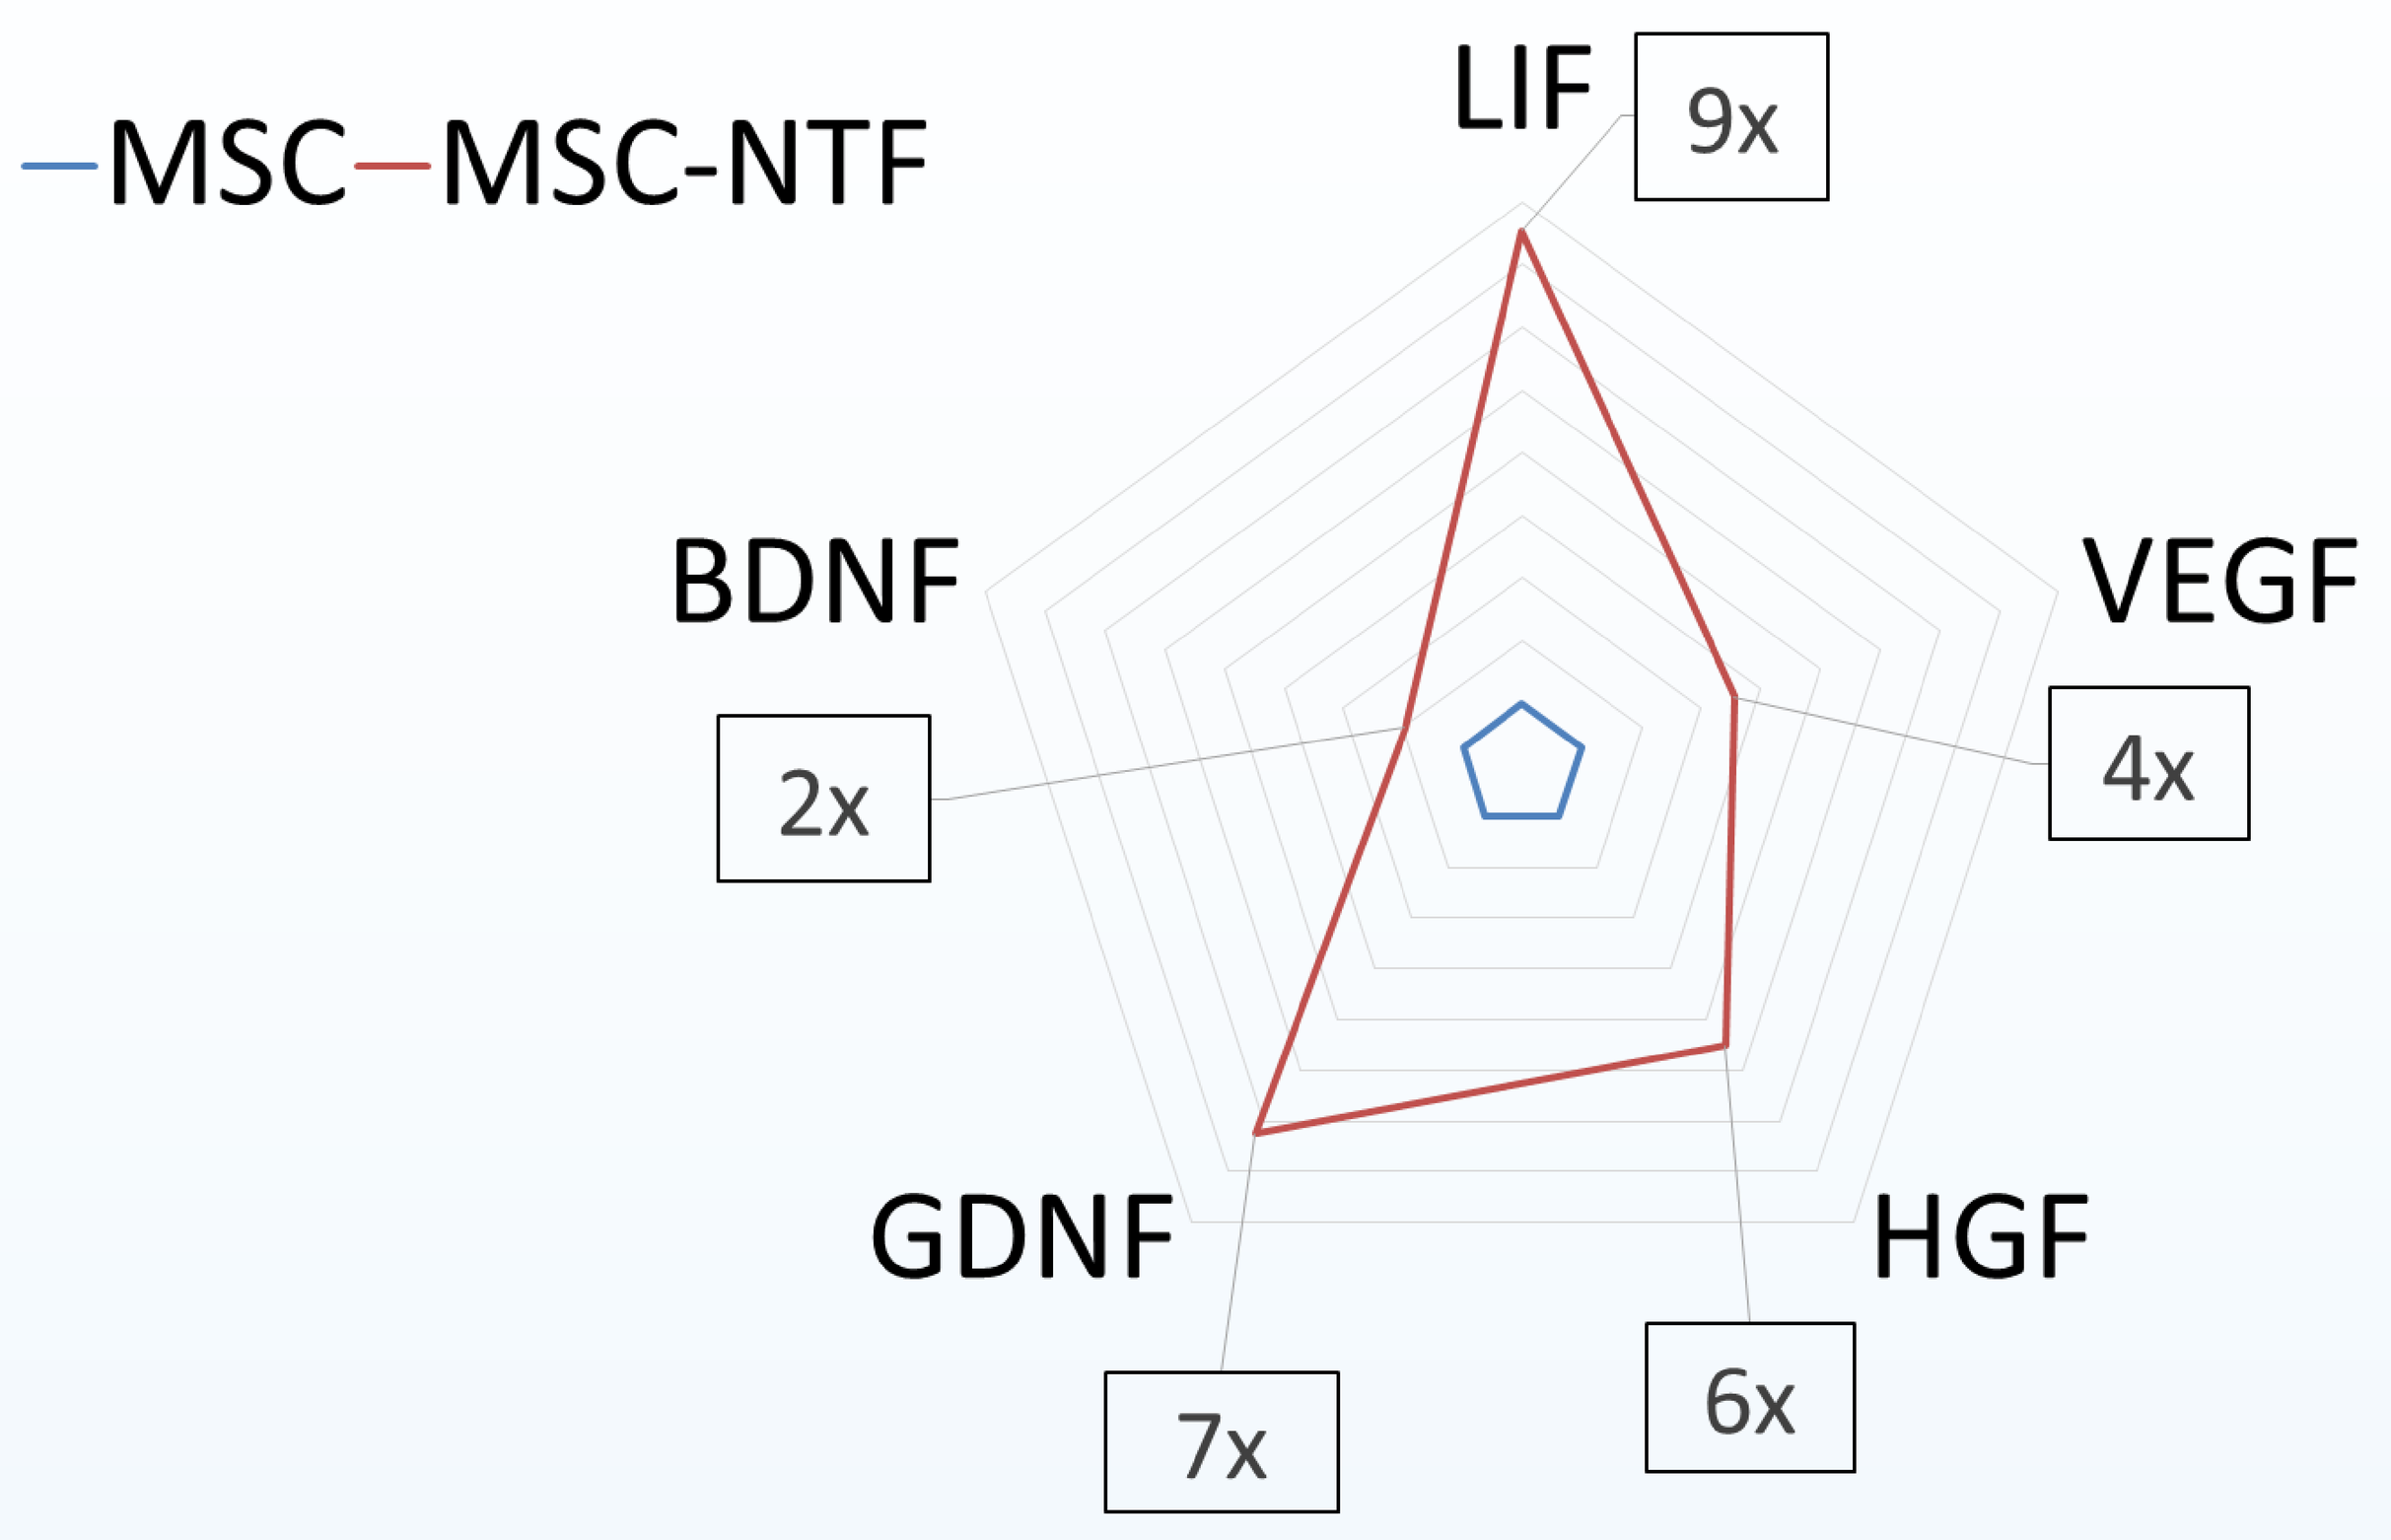
—MSC—MSC-NTF
LIF
9x
VEGF
4x
HGF
6x
GDNF
7x
BDNF
2x

A radar plot depicts the fold change in secretion of selected neurotrophic factors by MSC-NTF in comparison to MSC. Protein abundance was measured by ELISA. n=28 patients for all factors except LIF (n=18).

VEGF is intertwined with proteins which are upregulated in MSC-NTF

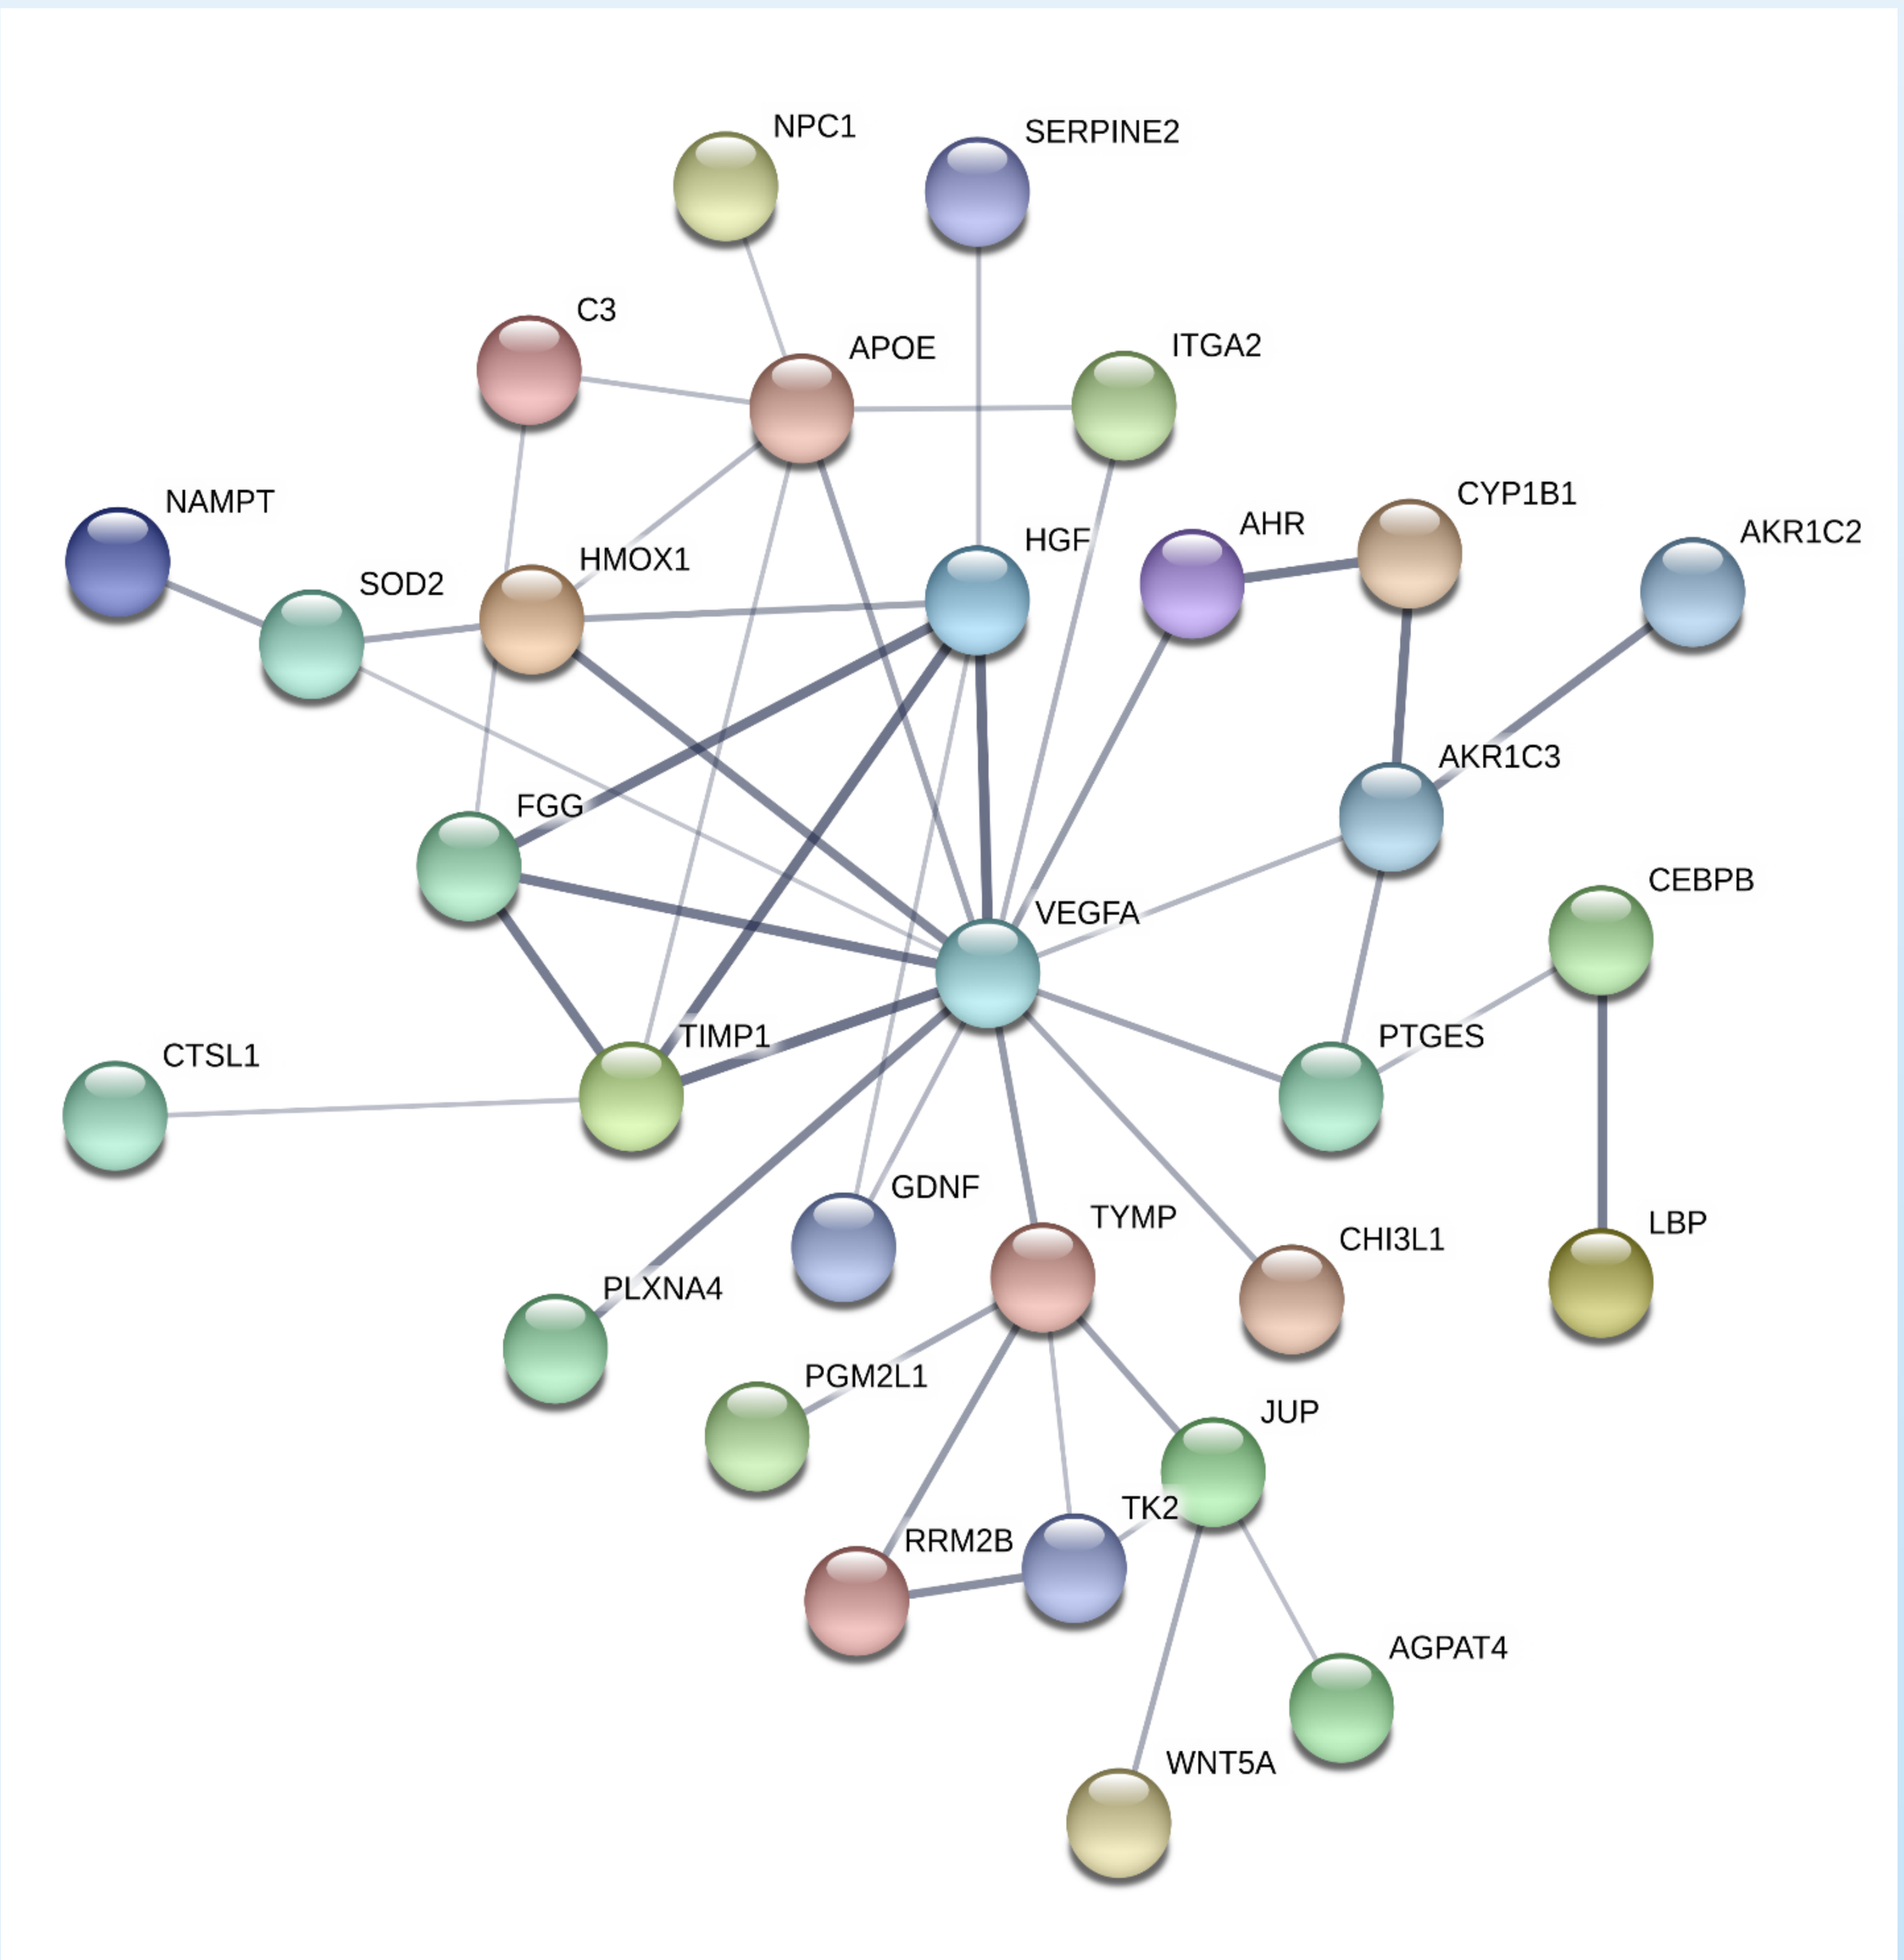
NPC1
SERPINE2
C3
APOE
ITGA2
NAMPT
SOD2
HMOX1
HGF
AHR
CYP1B1
AKR1C2
AKR1C3
FGG
VEGFA
CEBPB
CTSL1
TIMP1
PTGES
GDNF
TYMP
CHI3L1
LBP
PLXNA4
PGM2L1
JUP
RRM2B
TK2
AGPAT4
WNT5A

STRING analysis reveals interactions of VEGF with proteins which were found to be significantly upregulated in MSC-NTF vs. MSC by mass-spectrometry. Thicker line depicts stronger predicted interaction. Aryl hydrocarbon receptor (AHR) which was previously shown to upregulate VEGF expression was found to be significantly upregulated in MSC-NTF vs. MSC.

Changes in miRNA expression support increased VEGF secretion by MSC-NTF

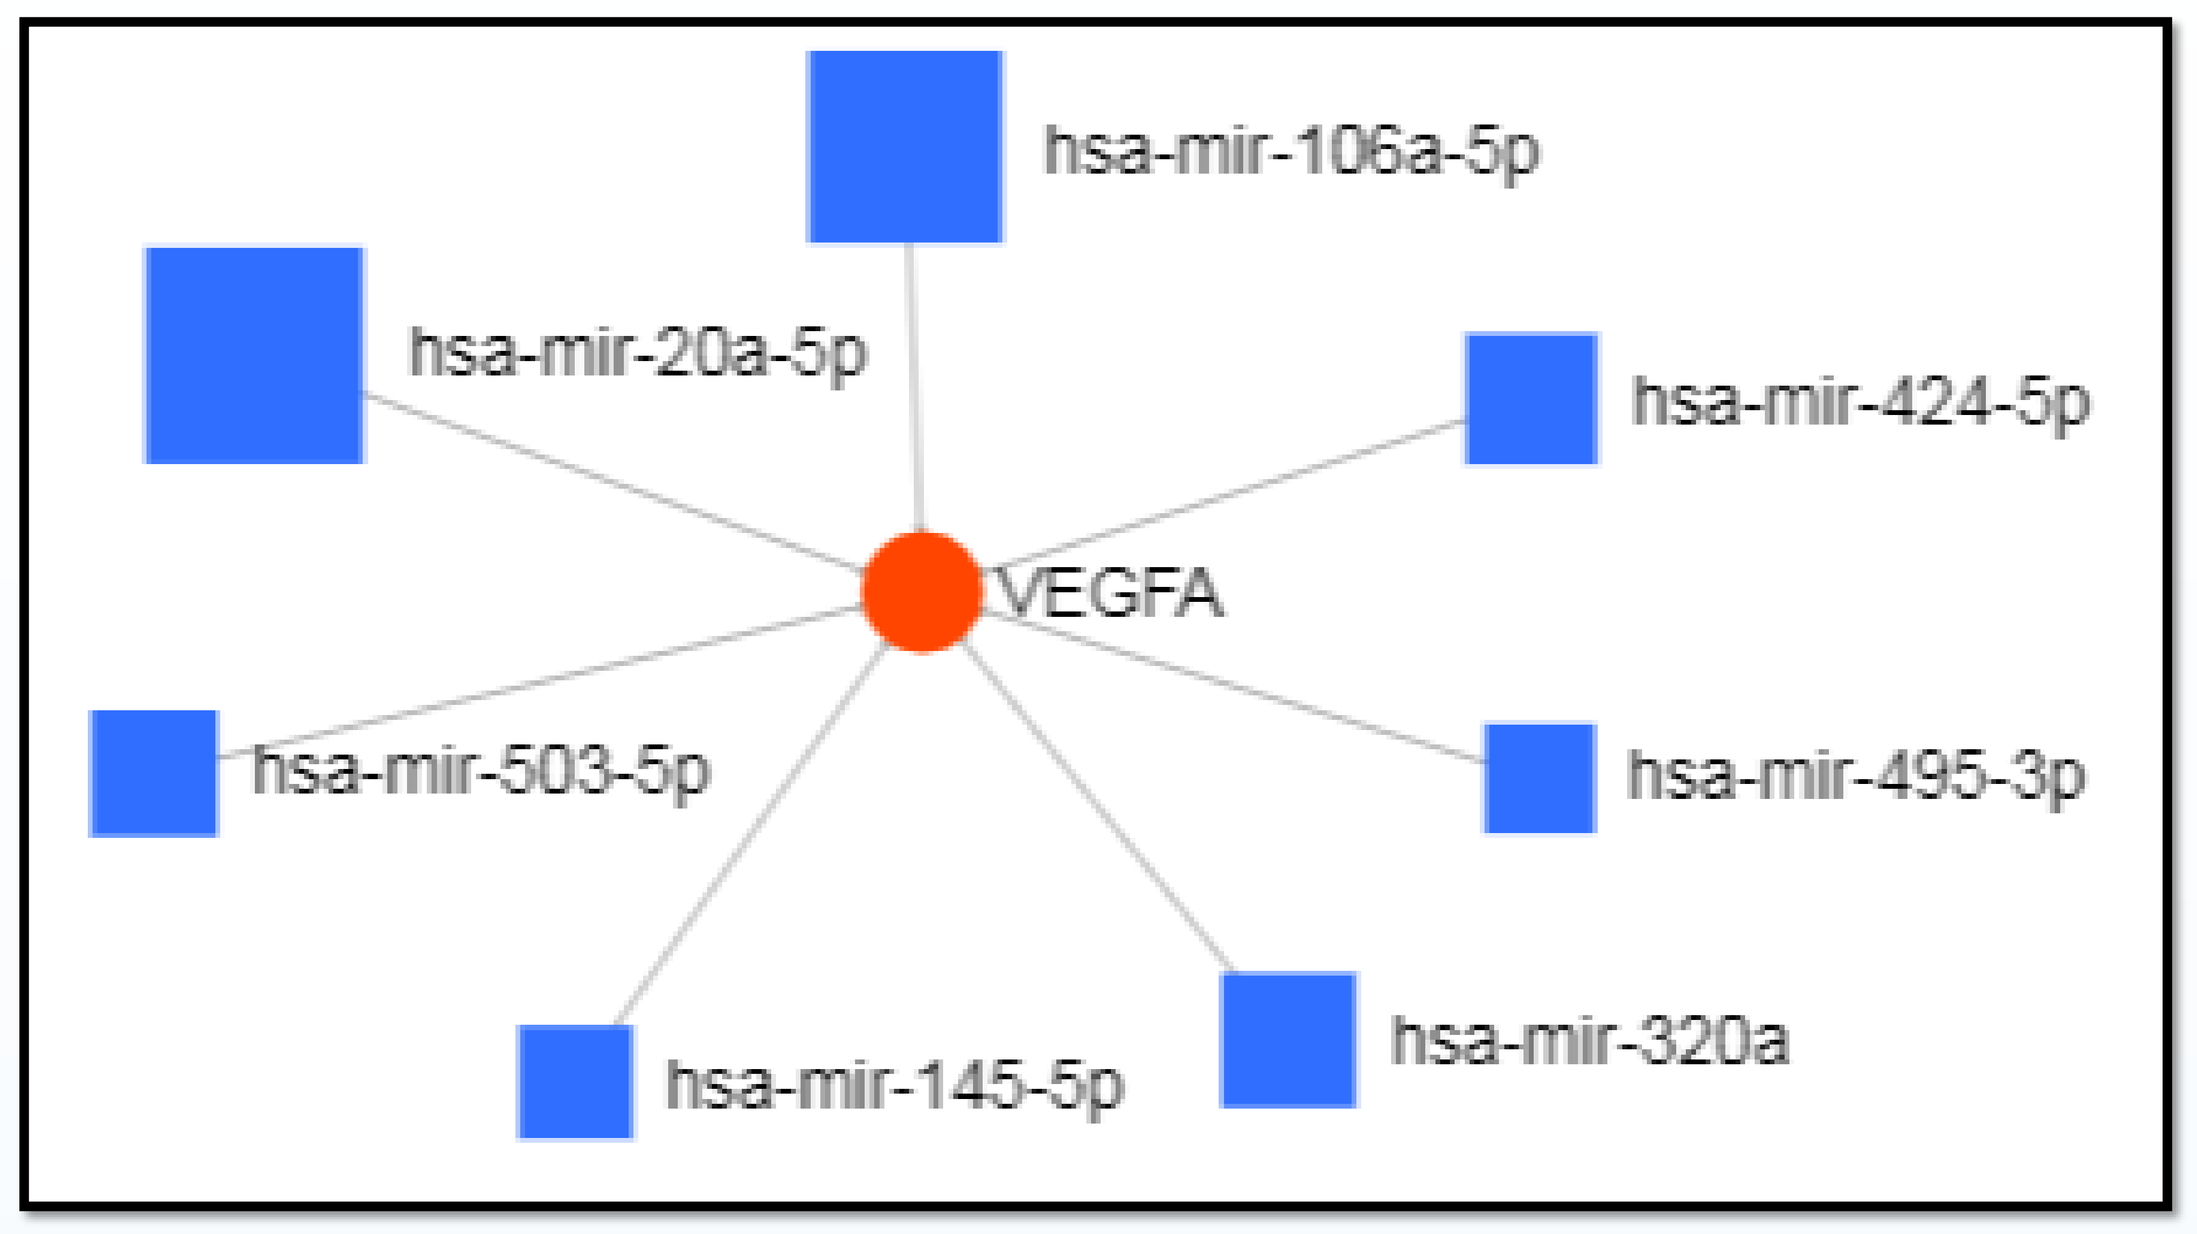
hsa-mir-106a-5p
hsa-mir-20a-5p
hsa-mir-424-5p
VEGFA
hsa-mir-503-5p
hsa-mir-495-3p
hsa-mir-145-5p
hsa-mir-320a

Twenty-two microRNAs were found to be significantly down regulated in MSC-NTFs vs. MSCs (pFDR<0.05, FC>1.5). Of these, seven microRNAs (depicted in the figure above) are predicted by the miRNet algorithm to target VEGF. Thus, down-regulation of these miRNAs should contribute to increased expression and secretion of VEGF.

Upregulation of VEGF in the CSF of patients treated with MSC-NTF

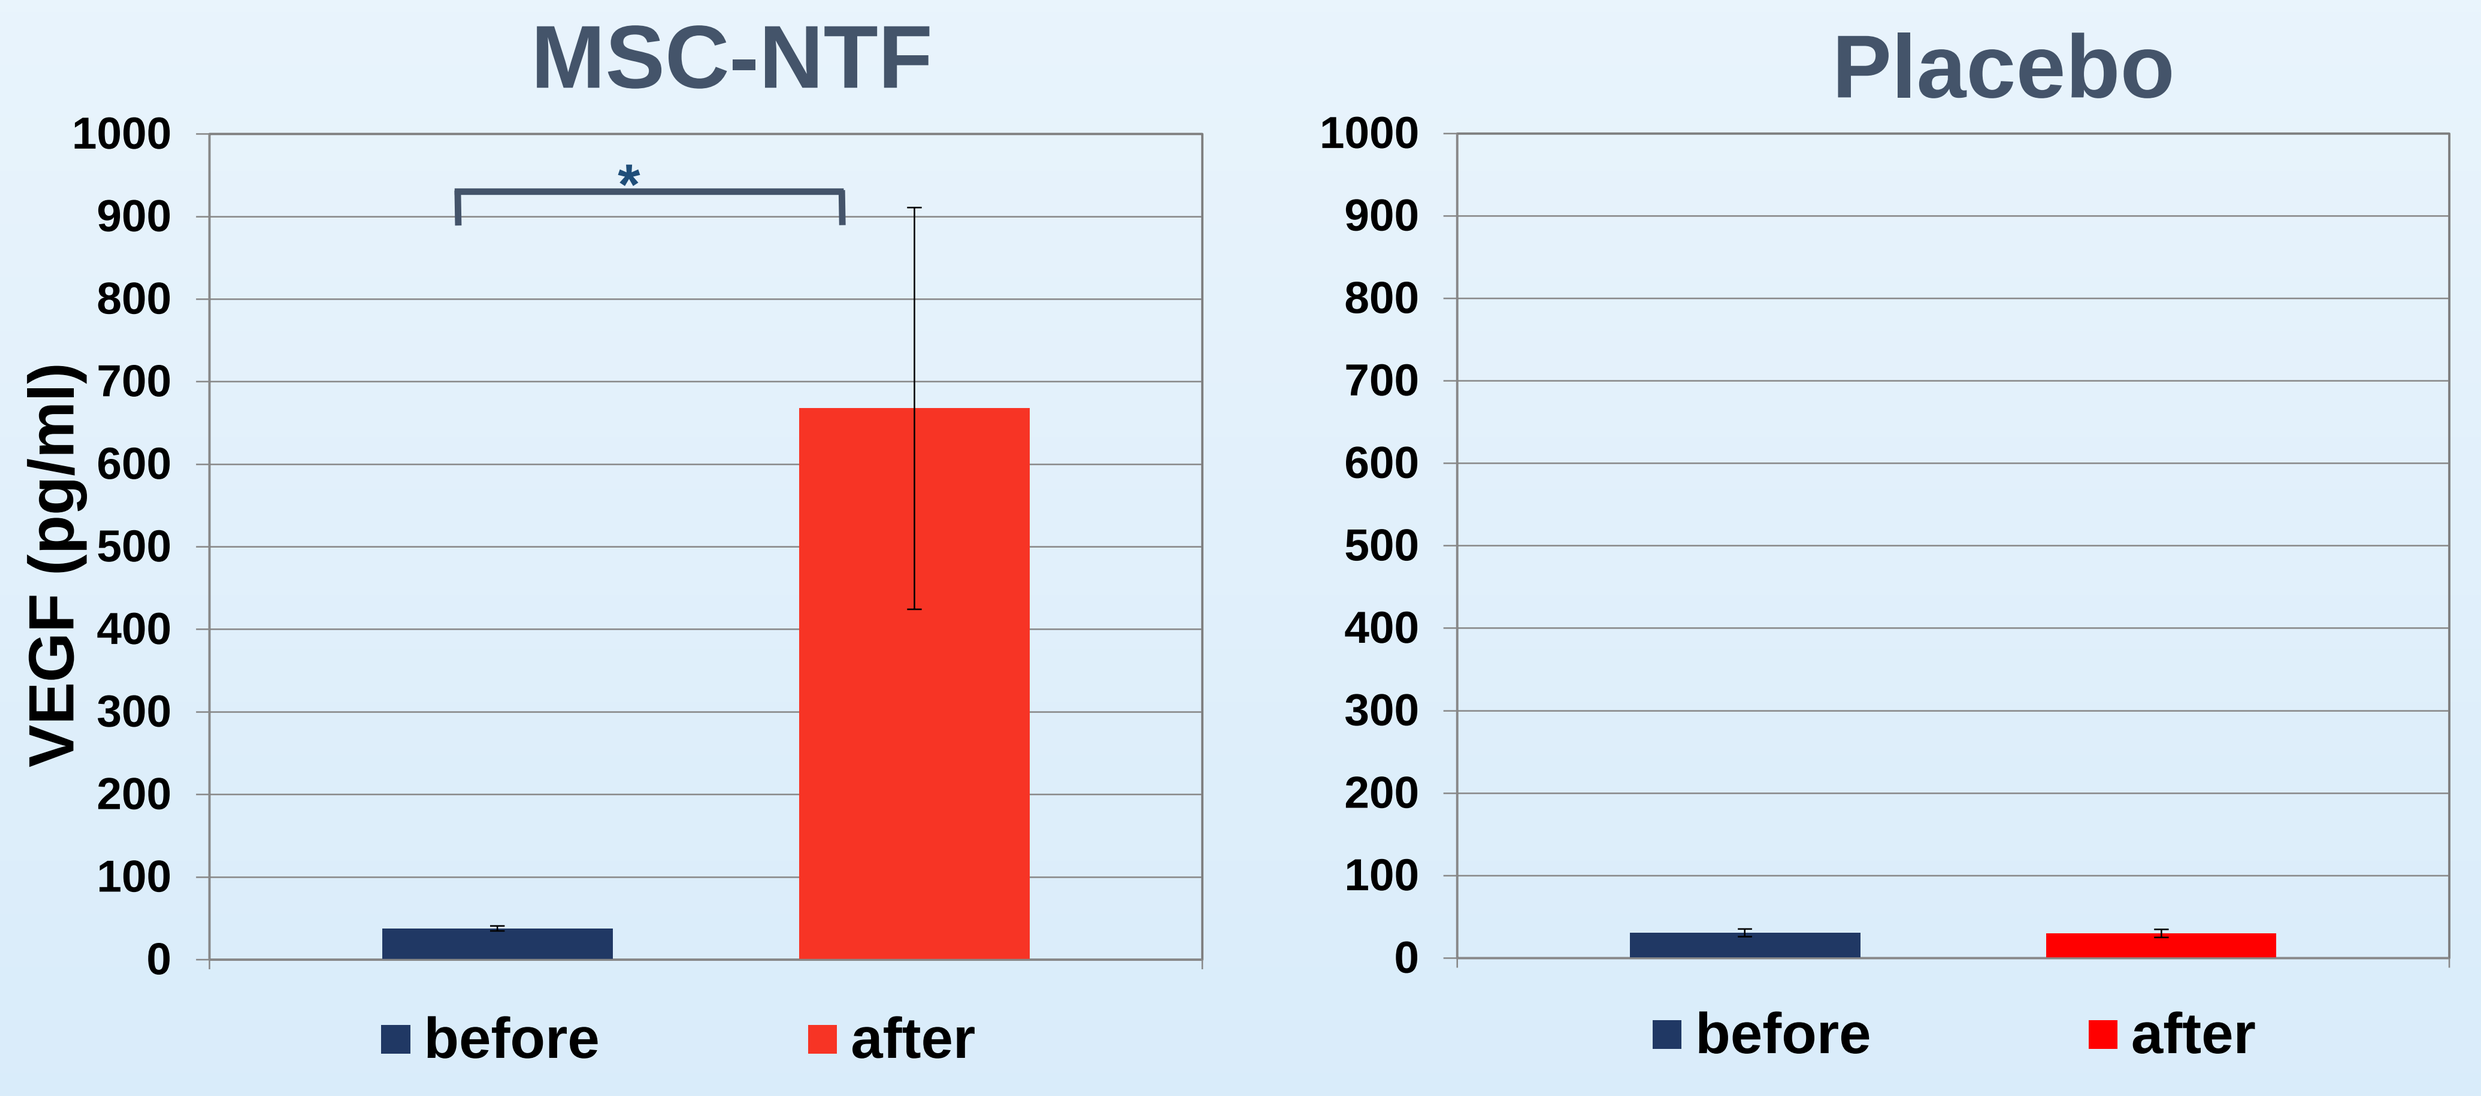
MSC-NTF
Placebo
VEGF (pg/ml)
1000
900
800
700
600
500
400
300
200
100
0
*
before
after

VEGF ELISA of CSF samples taken from patients before or two weeks after MSC-NTF/placebo intrathecal transfusion. (n=26 for MSC-NTF and n=9 for placebo, *p<0.05).

Conclusions

There are significant differences in protein and microRNA expression between MSC and MSC-NTF cells that may contribute to NurOwn®'s mechanism of action. Changes in microRNA expression are likely to contribute to increased VEGF secretion in MSC-NTF cells. The significantly increased secretion of VEGF and other NTFs by MSC-NTF cells may contribute to key ALS neuroprotective and immunomodulatory therapeutic mechanisms.